Recombinant IL-21 in Combination with Sorafenib as Second- or Third-Line Therapy for Metastatic Renal Cell Carcinoma (mRCC): Interim Results from a Phase 2 Study

Thomas Flaig[1], Brendan Curti[2], Michael Gordon[3], Peter VanVeldhuizen[4], Wilson Miller[5], Marc Ernstoff[6], Shailender Bhatia[7], Diana Hausman[8], Naomi Hunder[8], Nels Royer[8], John Thompson[7]

1 University of Colorado Health Sciences Center, Aurora, CO; 2 Providence Medical Center, Portland, OR; 3 Premiere Oncology of Arizona, Scottsdale, AZ; 4 University of Kansas Hospital Cancer Center, Kansas City, KS; 5 McGill University, Montreal, Quebec; 6 Norris Cotton Cancer Center, Lebanon, NH; 7 University of Washington/Seattle Cancer Care Alliance, Seattle, WA; 8 ZymoGenetics, Inc., Seattle, WA

INTRODUCTION

Treatment paradigms for metastatic renal cell carcinoma (RCC) have changed with the advent of targeted therapeutics, including the tyrosine kinase inhibitor sorafenib. Despite the clinical benefits obtained with targeted agents, durable responses are rare, and most subjects progress on treatment. Sequential therapy with other targeted agents is possible, although overall median progression-free survival (PFS) is decreased relative to the first-line setting. Everolimus, an inhibitor of mammalian target of rapamycin (mTOR), has demonstrated a median PFS of 4 months and a response rate of 1% in subjects with metastatic RCC whose disease had progressed on vascular endothelial growth factor (VEGF)-targeted therapy.[1] Sorafenib, administered to subjects with metastatic RCC refractory to either sunitinib or bevacizumab, is associated with a median PFS of 3.7 months and a 0% response rate.[2]

One approach to improve the outcomes obtained with targeted therapies in RCC is to administer them in combination with other active agents with a distinct mechanism of action. Interleukin-21 (IL-21), a member of the common gamma-chain cytokine family, enhances antitumor immunity through activation of cytotoxic T cells and natural killer (NK) cells. Unlike IL-2, IL-21 does not enhance proliferation of regulatory T cells that can limit antitumor autoimmunity. In Phase I clinical studies, single-agent recombinant IL-21 (rIL-21) had a safety profile compatible with outpatient administration and was associated with antitumor activity in RCC.[3,4] In preclinical studies, the combination of rIL-21 and sorafenib had additive effects on tumor shrinkage and prolonged survival in a murine RENCA model of RCC.

A Phase 1/2 study was initiated to evaluate the safety and antitumor effects of the combination of rIL-21 and sorafenib in subjects with metastatic RCC. Results from the completed Phase 1 dose-escalation portion of this study, conducted in subjects receiving first-, second-, or third-line therapy, have previously been reported, with a dose level of rIL-21 of 30 µg/kg identified as the maximum tolerated dose (MTD).[5] Interim results from the Phase 2 portion of this study, conducted in subjects receiving second- or third-line therapy after a prior targeted agent, are presented here.

1 Motzer RJ, Escudier B, Oudard S, et al, The Lancet 2008: 372:449-456
2 Shepard DR, Rini BI, Garcia JA, et al, J Clin Oncol, 2008 ASCO Annual Meeting Proceedings Vol 26, No. 15S (May 20 Supplement), 2008:5123
3 Davis ID, Skrumsager BK, Cebon J, et al, Clin Cancer Res 2007: 13(12):3630-3636
4 Thompson JA, Curti BD, Redman BG, et al, J Clin Oncol 2008: 26(12):2034-2039
5 Bhatia S, Curti BD, Gordon MS, et al, J Clin Oncol, 2008 ASCO Annual Meeting Proceedings Vol 26, No. 15S (May 20 Supplement), 2008:16008

STUDY DESIGN

This study was a Phase 1/2 open-label, dose-escalation study of rIL-21 administered in combination with the approved dose of sorafenib (400 mg po BID) in subjects with metastatic RCC

Phase 2

- Primary objective
 - Evaluate the safety of rIL-21 + sorafenib
- Secondary objectives
 - Evaluate anti-tumor activity of rIL-21 + sorafenib
 - Characterize pharmacokinetics of rIL-21 + sorafenib
- Endpoints
 - Safety (adverse events and lab abnormalities)
 - Objective response rate (ORR), assessed by investigator and independent review
- Subject population
 - Metastatic RCC, with history of prior nephrectomy and measurable disease per Recist
 - Receiving second- or third-line treatment after no more than 1 prior therapy with a regimen targeting the VEGF pathway, such as sunitinib, bevacizumab, or investigational agent targeting VEGF
 - Prior IL-2 therapy allowed
 - ECOG status 0 or 1

rIL-21 Treatment Schedule

Treatment Course (TC)=7 weeks; 30 µg/kg rIL-21[a] on Days 1-5 and 15-19; sorafenib 400 mg po BID daily[b]

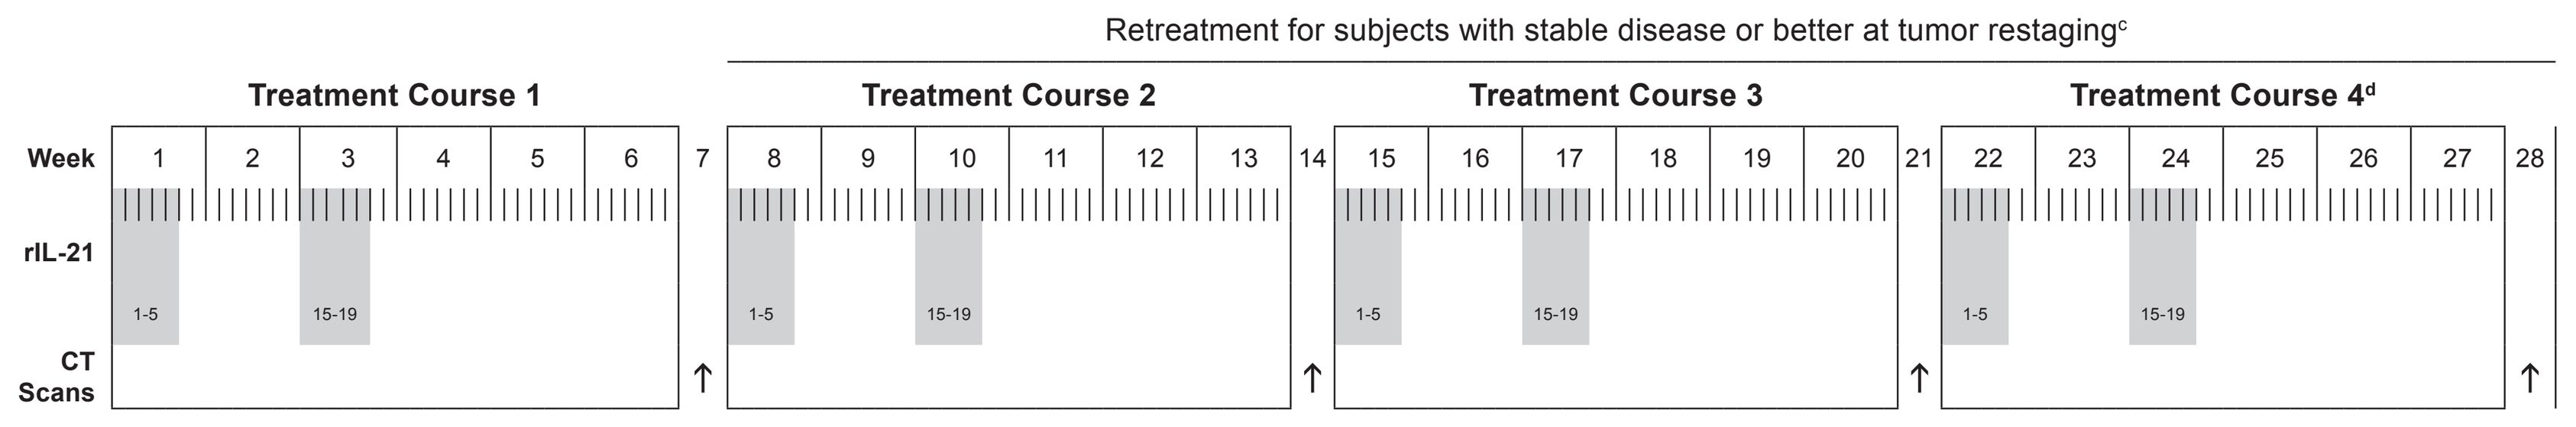
Retreatment for subjects with stable disease or better at tumor restaging[c]
Treatment Course 1
Treatment Course 2
Treatment Course 3
Treatment Course 4[d]
Week
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25 26 27 28
rIL-21
1-5
15-19
CT Scans

Note: Tumor restaging occurs during Week 7 of each treatment course
a Dose reductions of rIL-21 allowed in Treatment Course 2 and beyond
b Up to 2 dose reductions of sorafenib allowed for treatment-related toxicity any time on study
c Per investigator review
d Subjects with continued ongoing clinical benefit could continue to receive additional treatment courses

PHASE 2 RESULTS

- 33 subjects were enrolled in Phase 2 between January 2008 and August 2008
- The following data were available as of September 2008:
 - 32 subjects: safety data
 - 18 subjects: tumor response data per independent review

Subject Characteristics (N=32)[a]

Gender	
Male	26 (81%)
Female	6 (19%)
Age, median (range)	60 years (46–75)
ECOG performance status score	
0	15 (47%)
1	17 (53%)
Motzer risk[a]	
Favorable	16 (50%)
Intermediate	15 (47%)
Prior systemic treatments for mRCC	
Number[a]	
1	21 (66%)
2	10 (31%)
Agent	
Sunitinib	18 (56%)
IL-2	9 (28%)
Temsirolimus	2 (6%)
Pazopanib	1 (3%)
Other	11 (34%)

a Data unavailable for 1 subject

Subject Disposition by Treatment Course

- As of 30 Sept 2008, 18 of 33 subjects remain on study
 - 5 subjects discontinued due to adverse events
 - 9 subjects discontinued due to progressive disease[a]
 - 1 subject with SD discontinued at his request to receive retreatment with high-dose IL-2

	TC1	TC2	TC3	TC4	TC5
Number subjects starting treatment in each course	33	24	13	7	4
Completed with SD or better[b]	24	13	7	4	0
Still receiving treatment in TC	2	7	5	2	2
Withdrew during or at end of TC	7	4	1	1	2
Toxicity	3	2	0	0	0
PD[b]	4	1	1	1	2
Other reason	0	1	0	0	0

PD=progressive disease; SD=stable disease; TC=treatment course
a Per investigator assessment; includes subjects not yet evaluated by independent review
b Per investigator assessment only

Safety Observations

- Majority of AEs and laboratory abnormalities were Grade 1 or 2
- Grade 3 or higher AEs occurring in at least 10% of subjects included the following:
 - Hypophosphatemia (n=12)
 - Hand-foot syndrome (n=8)
 - Rash (n=5)
 - Thrombocytopenia (n=3)
- Serious adverse events considered at least possibly related to rIL-21 occurred in 6 subjects and included the following:
 - Acute renal failure (n=2)
 - Rash and dehydration (n=1)
 - Coagulopathy, metabolic acidosis, and neutropenia (with elevated creatinine) (n=1)
 - Dyspnea (n=1)
 - Acute hemolytic anemia (n=1)

Most Common Treatment-Emergent Adverse Events

	Any Grade[a] n (%)	≥ Grade 3 n (%)
ANY	32 (100%)	26 (81%)
Rash	20 (63%)	5 (16%)
Fatigue	18 (56%)	2 (6%)
Chills	16 (50%)	—
Hand-foot syndrome	16 (50%)	8 (25%)
Hypophosphatemia	16 (50%)	12 (38%)
Pyrexia	15 (47%)	—
Diarrhea	15 (47%)	2 (6%)
Vomiting	10 (31%)	1 (3%)
Flu-like illness	9 (28%)	—
Nausea	9 (28%)	1 (3%)
Alopecia	9 (28%)	—
Cough	7 (22%)	—
Headache	7 (22%)	—
Pruritus	7 (22%)	—
Hypocalcemia	6 (19%)	1 (3%)
Hyperhidrosis	5 (16%)	—
Hypotension	5 (16%)	—
Insomnia	5 (16%)	—
Pain of skin	5 (16%)	—
Thrombocytopenia	5 (16%)	3 (9%)

a Reported in 5 or more subjects

Most Common Treatment-Emergent Non-hematologic Lab Abnormalities (N=32)

	Any Grade[a] n (%)	≥ Grade 3 n (%)
↑ Triglycerides[b]	29 (94%)	2 (6%)
↓ Phosphate	26 (81%)	15 (47%)
↓ Albumin	24 (75%)	2 (6%)
↓ CO_2[b]	20 (65%)	1 (3%)
↓ Sodium[b]	19 (61%)	4 (13%)
↑ Bilirubin	19 (59%)	2 (6%)
↑ ALT	17 (53%)	1 (3%)
↑ AST	16 (50%)	3 (9%)
↓ Calcium	15 (47%)	—
↑ Magnesium[b]	12 (39%)	1 (3%)
↓ Magnesium[b]	11 (35%)	—
↑ Creatinine	9 (28%)	2 (6%)
↑ Uric acid[b]	7 (23%)	4 (13%)
↓ Potassium[b]	6 (19%)	1 (3%)

a Reported in 5 or more subjects
b N=31 evaluable subjects

Treatment-Emergent Hematologic Lab Abnormalities (N=32)

	Any Grade n (%)	≥ Grade 3 n (%)
↓ Lymphocytes	29 (91%)	20 (62%)
↓ Platelets	27 (84%)	7 (22%)
↓ Hemoglobin	22 (69%)	1 (3%)
↓ WBC	18 (56%)	4 (13%)
↓ Neutrophils	16 (50%)	4 (13%)

Sorafenib Dose Adjustments (N=32)

- Reduction of sorafenib dose required in 59% of subjects at some time on study

	n (%)
No reduction	10 (31%)
Dose reduced	19 (59%)
Discontinued due to toxicity	5 (16%) [a]
Discontinued due to other reason	1 (3%)

a Subjects who discontinued due to toxicity may have previously had dose reductions

Tumor Response

- As of Sept 2008, tumor response data per RECIST as assessed by independent review were available for 18 subjects
- Antitumor activity was observed, with disease control rate of 89% (16 of 18 evaluable subjects with best response of SD or better any time on study) by independent review
- 3 of 18 evaluated subjects (17%) with confirmed PR by independent review

Best Response per Recist at Any Time on Study By Independent Review (N=18)[a]

	n (%)
Disease control rate (CR + PR + SD)	16 (89%)
PR, confirmed	3 (17%)
PR, unconfirmed	2 (11%)
SD	11 (61%)
PD	2 (11%)

CR=complete response; PD=progressive disease; PR=partial response; SD=stable disease
a Snapshot of data as of Sept 2008; treatment and assessment of tumor response is ongoing

Maximum Tumor Reduction on Study By Independent Review (N=17)[a]

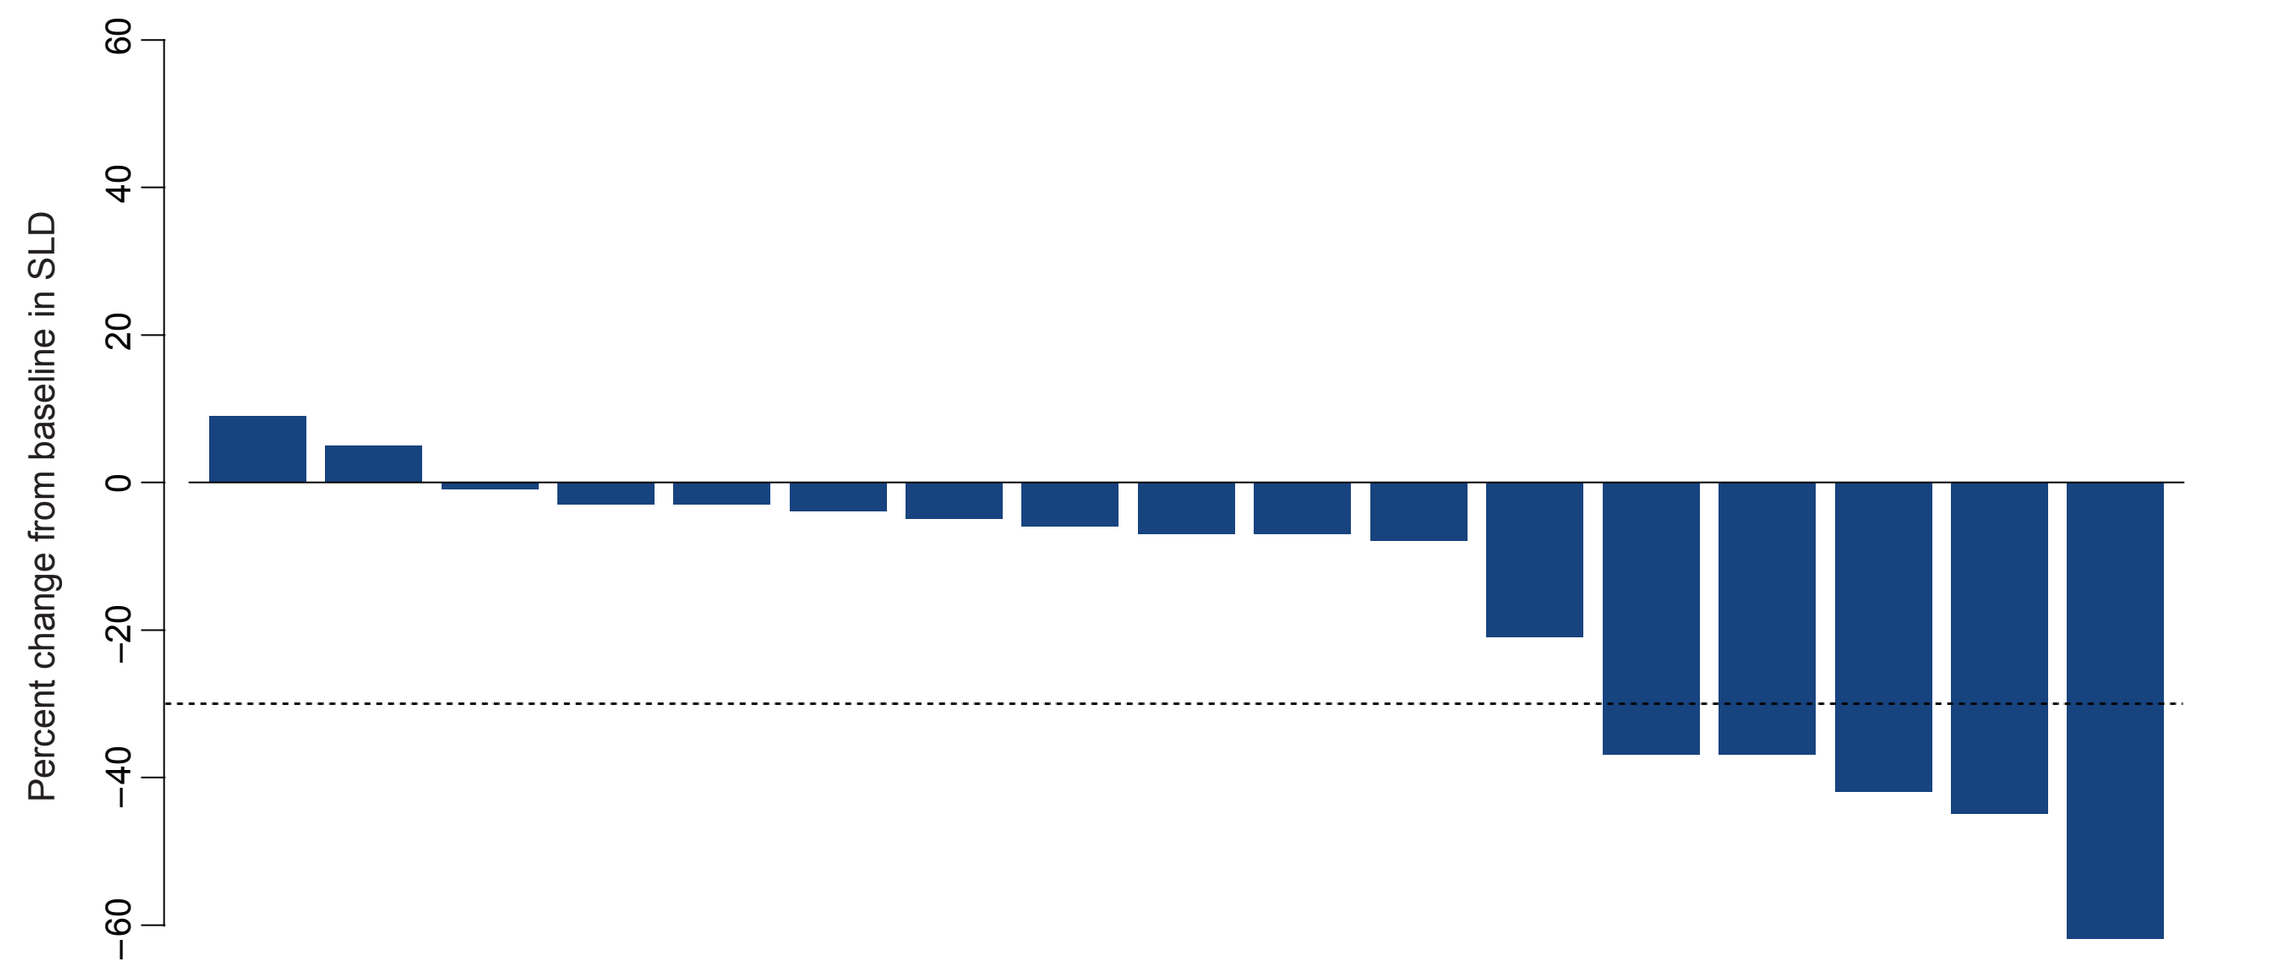
Percent change from baseline in SLD
60
40
20
0
−20
−40
−60

SLD=sum of the longest diameters of target lesions
Data presented for subjects who were evaluated by independent review
a One subject had no percent change recorded and is therefore not represented in diagram

Duration of Disease Control (N=33)[a]

- Median PFS has not yet been determined
 - 14 subjects still receiving treatment in Treatment Course 1, 2, or 3
- Data are available through Treatment Course 3 (equivalent to at least 21 weeks on study) for 19 subjects
 - 7 subjects completed Treatment Course 3 with SD or better[b]
 - 6 subjects had PD prior to end of Treatment Course 3[b]
 - 5 subjects discontinued due to toxicity prior to end of Treatment Course 3
 - 1 subject withdrew with SD at the end of Treatment Course 2

a As of 30 Sept 2008; data presented from enrolled subjects
b Per investigator assessment

ABSTRACT

Background Despite the positive impact of tyrosine kinase inhibitors (TKIs) and mTor inhibitors on the outcome for mRCC, complete responses are rare and long-term survival remains poor. Recombinant IL-21 (rIL-21), a cytokine that enhances CD8+ T cell and NK cell activity, has single-agent anti-tumor activity as shown in Phase 1 studies. The combination of rIL-21 plus the TKI sorafenib was tested in a Phase 1 study in the outpatient setting. We now report interim results of a Phase 2 study to evaluate the safety, pharmacokinetics, and anti-tumor efficacy of rIL-21 plus sorafenib using the maximum tolerated dose determined in Phase 1.

Methods 33 subjects with mRCC will be enrolled from 16 sites in the United States and Canada to receive second or third-line therapy with sorafenib 400 mg BID plus 30 µg/kg rIL-21 IV on days 1–5 and 15–19 of each 6-week treatment course. Tumor response per Recist criteria will be assessed by the investigator and by independent radiologic review.

Results As of 19 May 2008, 18 subjects were treated; the first 15 are summarized here. Median age was 59 (range 47–75), male:female ratio was 11:4, and ECOG performance status was 0 (n=6) or 1 (n=9). Subjects had received 1 (n=10) or 2 (n=5) previous lines of therapy, which included sunitinib (n=11), temsirolimus (n=3), IL-2 (n=3), pazopanib (n=1), everolimus + avastin (n=1), and vinblastine + interferon (n=1). Most adverse events (AEs) were Grade 1 or 2, and consistent with the known toxicity of rIL-21 and sorafenib. Common AEs (> 20% of subjects) included rash, diarrhea, hand-foot syndrome, flu-like illness, fever, chills, and pruritus. Grade 3 AEs occurred in 7 subjects and were hand-foot syndrome (n=3), neutropenia (n=2), thrombocytopenia (n=2), rash (n=1), elevated liver function tests (n=1), metabolic acidosis (n=1), coagulopathy (n=1), and acute renal failure (n=1). All 9 subjects for whom tumor assessment is available had stable disease, with tumor shrinkage of 0-27% as measured by the investigator. Seven of 10 subjects who have completed the 1st treatment course have received additional courses; none have discontinued therapy due to progressive disease.

Conclusions Outpatient therapy with rIL-21 plus sorafenib is well tolerated with appropriate dose modification and associated with anti-tumor activity as a second- or third-line therapy for mRCC. Updated results from all available subjects in Phase 2, including 6 months of follow-up for the first 15 subjects, will be available at the meeting.

CONCLUSIONS

- **In the second- and third-line setting for metastatic RCC, combination treatment with rIL-21 and sorafenib is associated with an ORR and disease control rate that compare favorably to results obtained with single-agent TKIs and mTOR inhibitors after failure of initial therapy**
 - **ORR of 17% (3 of 18 subjects with confirmed PR by independent review) suggests potential additive effect of rIL-21**
 - **While median PFS cannot yet be determined, 7 subjects have completed at least 3 treatment courses with SD or better (equivalent to minimum duration of PFS of 21 weeks)**
- **Toxicity of combination therapy is manageable in outpatient setting, although the majority of subjects (59%) required dose reductions of sorafenib, and careful attention to fluid status, renal function, and dermatologic adverse events is required**

ZymoGenetics
EORTC-NCI-AACR Symposium, October 2008, Geneva, Switzerland